Preclinical Development of ASC-618, an Advanced Human Factor VIII Gene Therapy Vector for the Treatment of Hemophilia A: Results from FRG-KO Humanized Liver Mice, C57Bl/6 Mice and Cynomolgus Monkeys

ASC Therapeutics

Milena Veselinovic[1], Avital Gilam[1], Adam Ross[1], Chengtao Yang[1], Yin Zhang[1], Tanushree Jaitly[1], Vladimir Pak[1], Aanal Bhatt[1], Mehma Kaur[1], Eliza Dewangan[1], Tanvee Sawant[1], Gil Gonen-Yaacovi[1], Lijing Li[1], Gabriela Denning[2], H. Trent Spencer[2], Christopher B Doering[2], Ling-jie Kong[1], Ruby Yanru Chen-Tsai[3], Ruhong Jiang[1], H. Steve Zhang[1], Zoya Gluzman- Poltorak[1]

[1]ASC Therapeutics, Milpitas, CA,[2]Expression Therapeutics, LLC, Tucker, GA,[3]ASC, Milpitas, CA

Abstract

Although several AAV-based gene therapies for hemophilia A are currently under evaluation in clinical trials, there is still an unmet medical need for different AAV serotypes, more efficient transgene vectors and reduced AAV doses to achieve high and sustained factor VIII expression with milder immunosuppressive treatments. ASC-618 is an advanced recombinant AAV2/8 vector with the shortest vector genome compared to other gene therapy constructs that have been tested in the clinic. It encodes a liver specific codon optimized (LCO) bioengineered B-domain deleted hFVIII (ET3) under a synthetic Hepatic Combinatorial Bundle (HCB) promoter (HCB-ET3-LCO). The HCB-ET3-LCO construct was previously characterized by Expression Therapeutics/Emory University in hemophilia A murine model and licensed in for further therapeutic development at ASC Therapeutics. Compared to the standard hFVIII transgene (HSQ), used in most hemophilia A gene therapies, ET3 demonstrates 10-100 fold enhancement in biosynthesis, and thus significantly better therapeutic potential. Here we present the results of safety and efficacy experiments comparing ET3 with HSQ in three species, namely C57Bl/6 murine model, cynomolgus monkeys and humanized liver mouse model (FRG-KO). In all 3 species, higher levels of circulating therapeutic product FVIII were seen in animals treated with AAV2/8 HCB-ET3-LCO, compared to AAV2/8 HCB-HSQ-LCO vector. In the C57Bl/6 murine pilot study, stable human FVIII expression was detected by human FVIII specific ELISA, with highest mean ET3 FVIII levels reaching around 50% (0.5 IU/mL), 300% (3 IU/mL) and 350% (3.5 IU/mL) of normal levels at the 5E10, 5E11, and 5E12 vg/kg dose, respectively. The highest mean HSQ FVIII expression levels were 7-fold and 3-fold lower at the 5E11 and 5E12 vg/kg dose, respectively, while at 5E10 vg/kg dose no HSQ expression was detected. The same trend of higher expression levels of ET3 was observed in the cynomolgus monkey study, with ~ 30% (0.3 IU/mL) of normal levels at the 5E11 vg/kg dose of AAV2/8 HCB-ET3-LCO. Importantly, when both vectors were tested in the humanized liver FRG-KO model at the 3E12 vg/kg dose level, the mean human FVIII levels reached around 480% (4.8 IU/mL) of normal levels after ET3 treatment while after HSQ treatment they only reached around 30%. Moreover, RNAscope analysis of liver tissue in FRG-KO model confirmed that ASC-618 drove high ET3 mRNA level expression in human hepatocytes. Safety studies conducted in all three models, including clinical observations, food consumption monitoring, body weight and temperature, liver enzyme and gross pathology evaluation, showed no toxicity effects. Together, these results demonstrate that ASC-618 is well-tolerated in animal models and has the potential of providing therapeutic benefit to patients at reduced vector doses.

ASC-618 Gene Therapy – AAV8 HCB-ET3-LCO – Key Advantages

ASC-618 Transgene and Expression Cassette Design

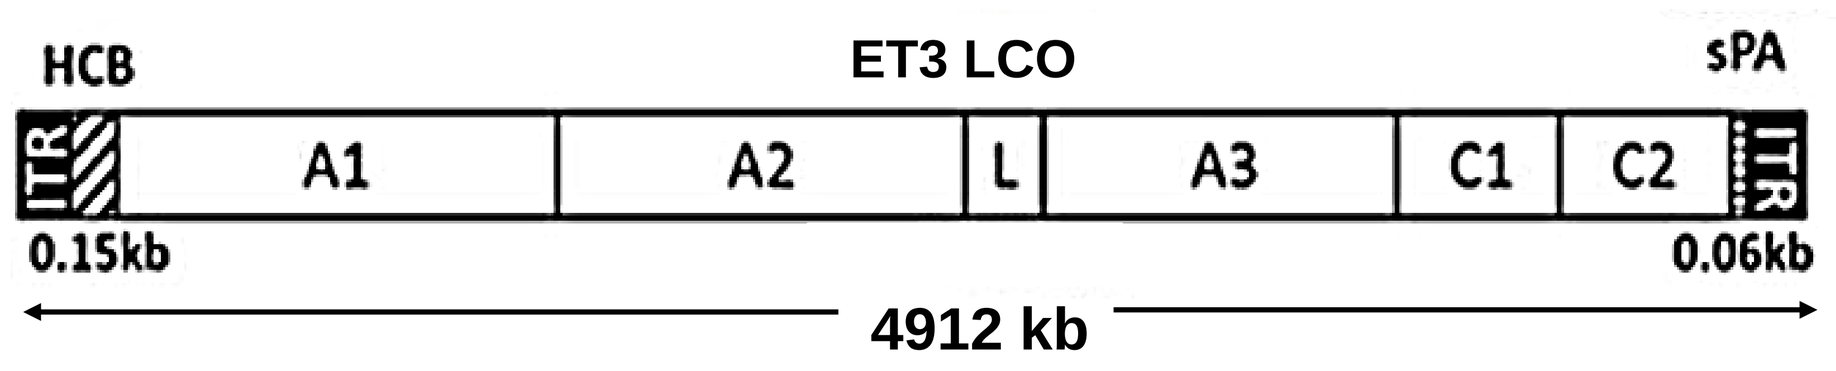

HCB
ET3 LCO
sPA
ITR
A1
A2
L
A3
C1
C2
ITR
0.15kb
0.06kb
4912 kb

Fig 1. AAV2(ITR)/AAV8 HCB-ET3-LCO design

1. **HCB promoter** *(Brown HC et al.Mol Ther Methods Clin Dev. Jan 2018)*

- Liver-directed promoter minimizes the packaging size and allows for higher protein expression levels

2. **ET3 transgene** *(Brown HC et al. Mol Ther Methods Clin Dev. Aug 2014)*

- ET3 is bioengineered human B domain deleted (BDD) fVIII with porcine A1 and A3 domain elements
- 10 –100x higher biosynthesis, expression & secretion vs. BDD human fVIII (HSQ)
- Greater stability than BDD human fVIII following thrombin activation
- Comparable reactivity with human fVIII inhibitors to human BDD fVIII constructs = indistinguishable nonclinical immunogenicity

3. **Liver codon optimization** *(Brown HC et al. Mol Ther Methods Clin Dev. Jan 2018)*

- Liver-Codon Optimization (LCO) improves transgene expression levels

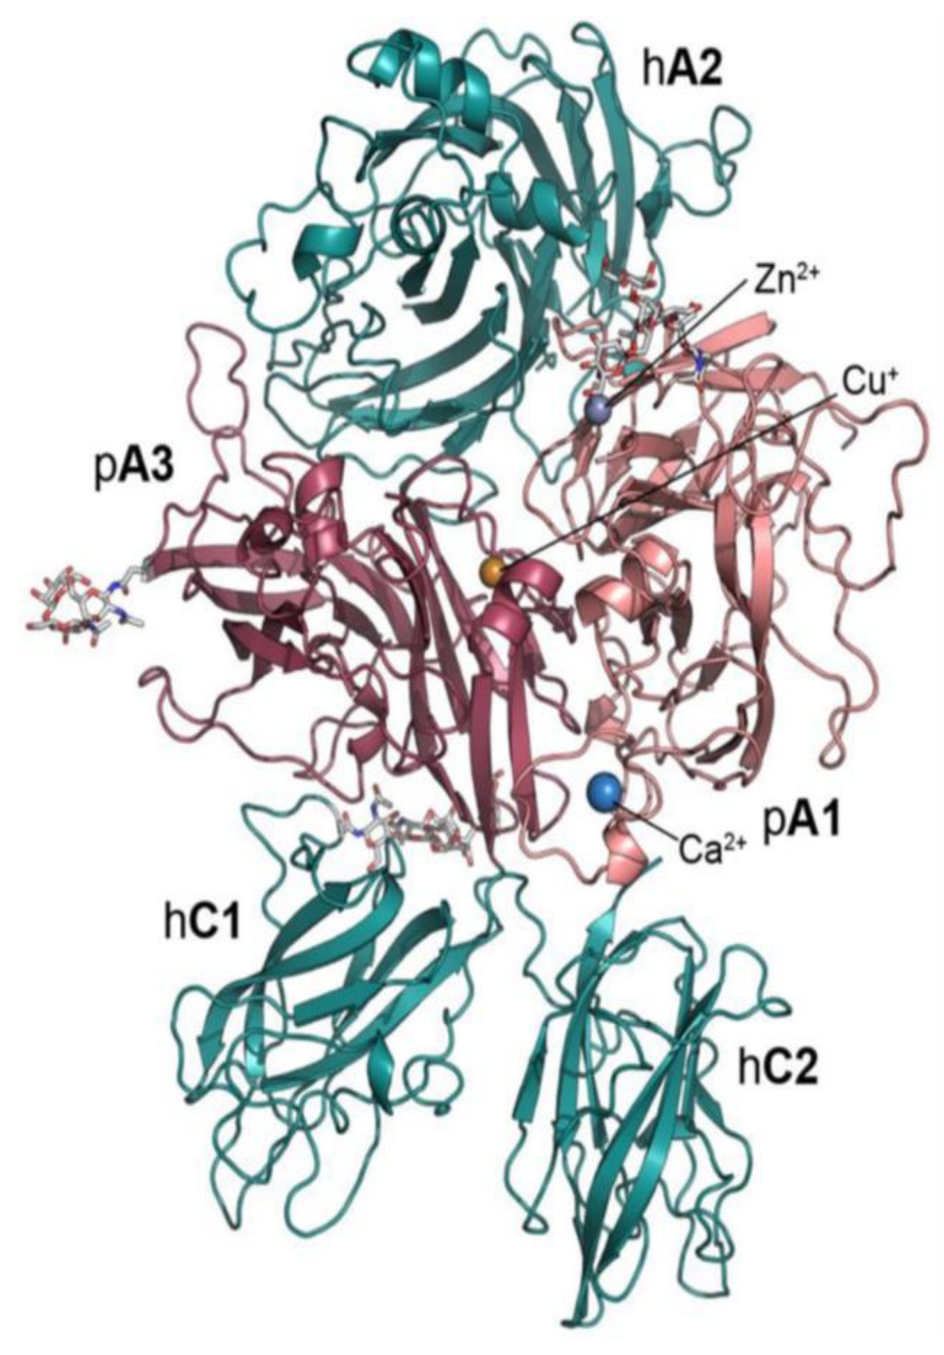

hA2
Zn2+
Cu+
pA3
pA1
Ca2+
hC1
hC2

Fig 2. The structure of bioengineered ET3 *(Smith IW et al. J Thromb Haemost. Jan 2020)*

ASC-618 Efficacy In Vivo - Humanized Liver FRGKO model

ET3/HSQ protein levels by ELISA

Liver infectivity and ET3/HSQ mRNA expression

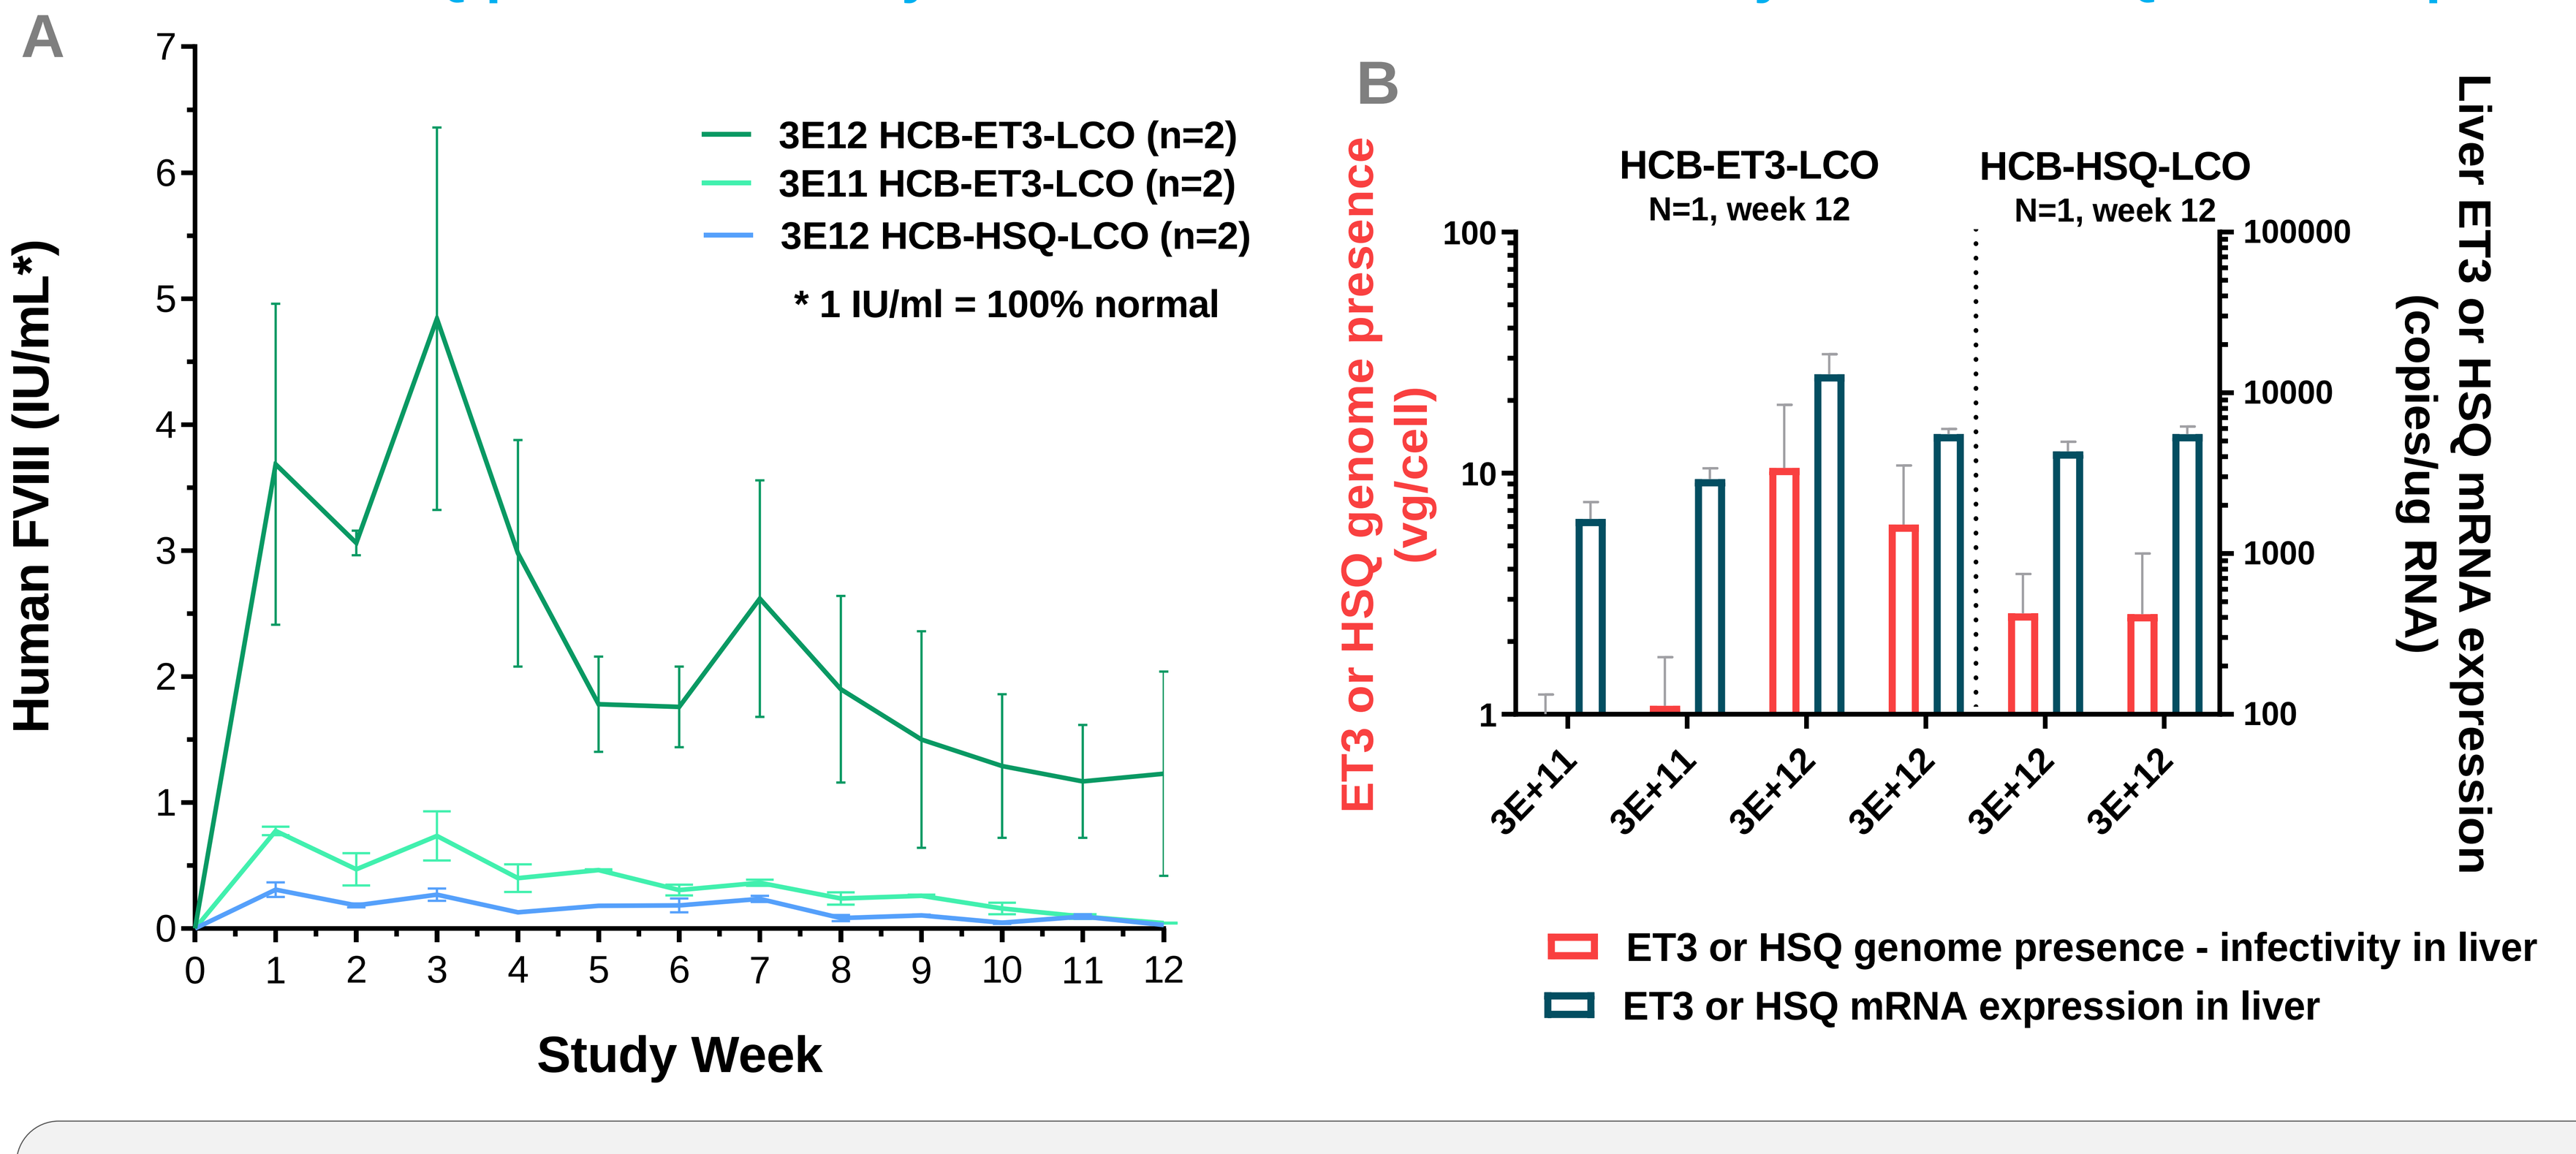

A
Human FVIII (IU/mL*)
3E12 HCB-ET3-LCO (n=2)
3E11 HCB-ET3-LCO (n=2)
3E12 HCB-HSQ-LCO (n=2)
* 1 IU/ml = 100% normal
Study Week
B
HCB-ET3-LCO
N=1, week 12
HCB-HSQ-LCO
N=1, week 12
ET3 or HSQ genome presence (vg/cell)
Liver ET3 or HSQ mRNA expression (copies/ug RNA)
3E+11
3E+11
3E+12
3E+12
3E+12
3E+12
ET3 or HSQ genome presence - infectivity in liver
ET3 or HSQ mRNA expression in liver

Fig 3. A. Human fVIII ELISA – circulating protein levels in FRGKO plasma (IU/mL; mean, SEM). HCB-ET3-LCO doses tested were 3E11 vg/kg and 3E12 vg/kg, while HCB-HSQ-LCO was tested at 3E12 vg/kg (IV injection, n=2). All groups were followed up to w12. **B.** In vivo infectivity - ET3 or HSQ genome presence in liver tissue (ddPCR, vg/cell – 4 lobe mean, SD), ET3 or HSQ mRNA expression in liver tissue (ddPCR, copy number/µg RNA – 4 lobe mean, SD).

Summary: ET3 showed higher protein levels at the same 3E12 vg/kg dose, while mRNA expression levels were comparable in the liver for both constructs. As expected, liver infectivity and mRNA expression were lower for ET3 at 3E11 vg/kg vs. 3E12 vg/kg HSQ group, while plasma ET3 protein levels were higher, indicating enhanced ET3 protein biosynthesis and secretion from human hepatocytes

High level of ET3 mRNA expression in human hepatocytes by RNAscope

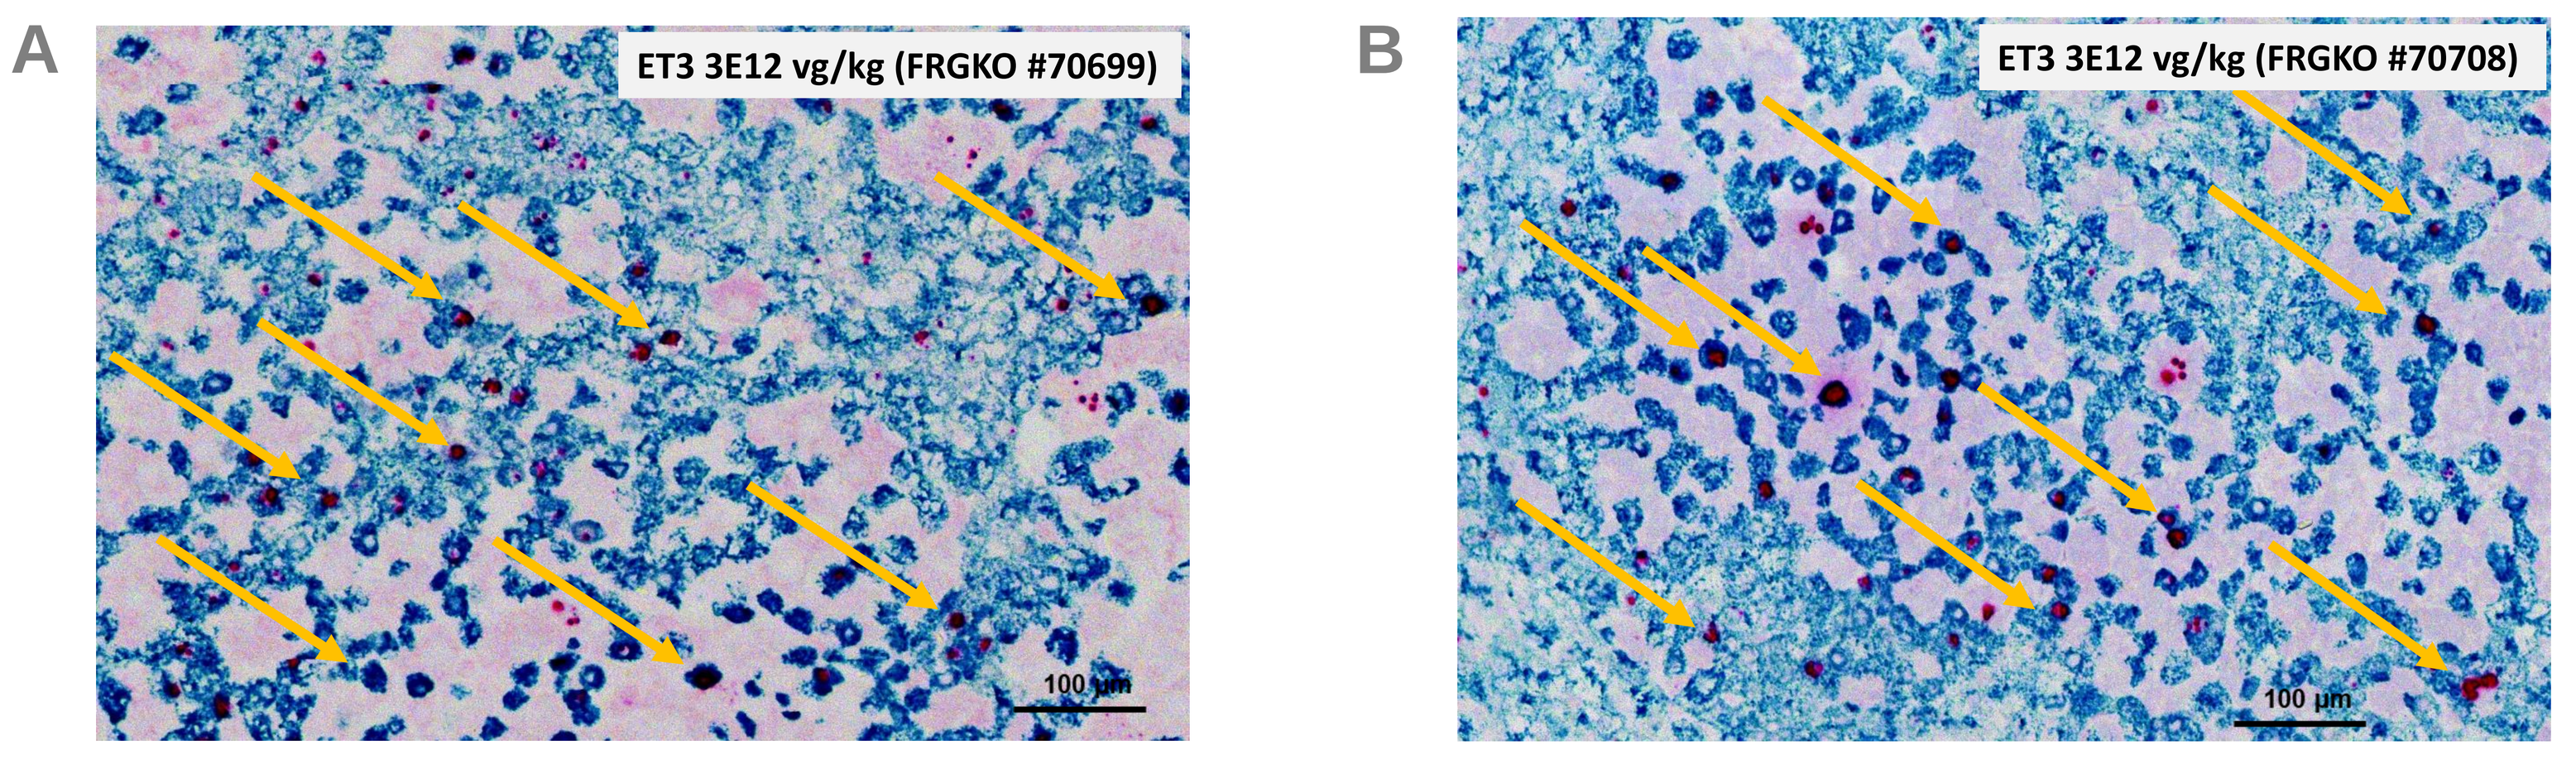

A
ET3 3E12 vg/kg (FRGKO #70699)
B
ET3 3E12 vg/kg (FRGKO #70708)
100 µm

Fig 4. A,B. ET3 fVIII mRNA tissue distribution by liver RNAscope – **representative images from 2 FRGKO mice shown. ET3 mRNA production was confirmed in ~ 50% human hepatocytes at 3E12 vg/kg dose, and resulted in therapeutically relevant plasma levels of ET3 fVIII.**
Red – ET3 fVIII mRNA; Blue – human albumin mRNA; Yellow arrows – ET3 mRNA in human hepatocytes

ASC-618 Efficacy In Vivo – C57Bl/6 Murine Model

ET3/HSQ protein levels by ELISA

ET3 vs. HSQ FVIII protein level comparison (5E11vg/kg dose)

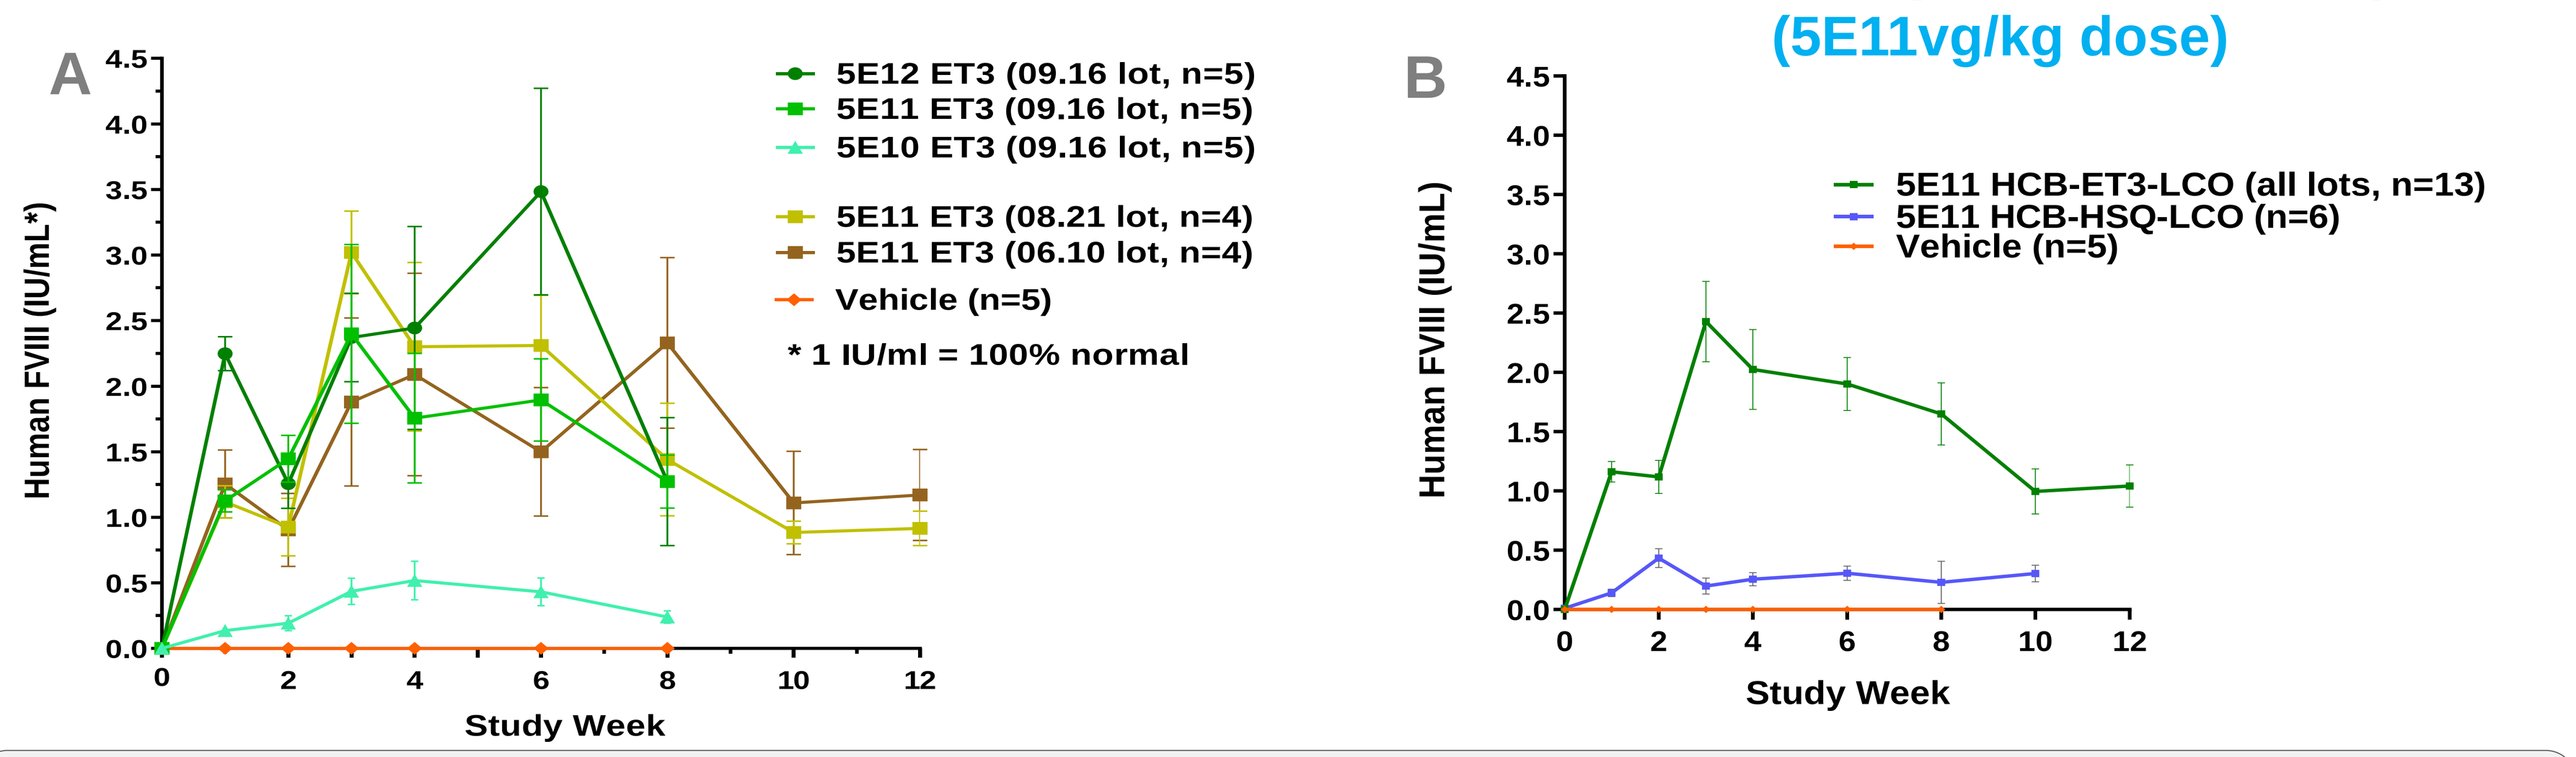

A
Human FVIII (IU/mL*)
5E12 ET3 (09.16 lot, n=5)
5E11 ET3 (09.16 lot, n=5)
5E10 ET3 (09.16 lot, n=5)
5E11 ET3 (08.21 lot, n=4)
5E11 ET3 (06.10 lot, n=4)
Vehicle (n=5)
* 1 IU/ml = 100% normal
Study Week
B
Human FVIII (IU/mL)
5E11 HCB-ET3-LCO (all lots, n=13)
5E11 HCB-HSQ-LCO (n=6)
Vehicle (n=5)
Study Week

Fig 5. A. Human fVIII ELISA – circulating protein concentration in C57Bl/6 plasma (IU/mL; mean, SEM). HCB-ET3-LCO was tested at 3 dose levels – 5E10, 5E11 and 5E12 vg/kg (IV injection, n= 4 or 5). Three mid dose groups (5E11 vg/kg) were included to test response consistency between different virus production lots. Two of the mid dose groups were followed up to w12, while all other study groups were followed up to w8. **B.** ET3 fVIII protein levels compared to HSQ protein levels at 5E11 vg/kg dose in C57Bl/6 murine plasma. 5E11 vg/kg groups from HCB-ET3-LCO study shown in A are compared to HCB-HSQ-LCO dosed animals from a separate C57Bl/6 study.
Summary: Stable and robust human fVIII expression was detected by ELISA, with highest mean ET3 fVIII levels reaching around 50% (0.5 IU/mL), 300% (3 IU/mL) and 350% (3.5 IU/mL) of normal levels at the 5E10, 5E11, and 5E12 vg/kg dose, respectively. ET3-treated animals showed improved protein bioproduction vs. HSQ, at multiple tested dose levels (5B for 5E11vg/kg, data not shown - other doses).

Liver infectivity and ET3/HSQ mRNA expression

ET3 mRNA Biodistribution in tissues

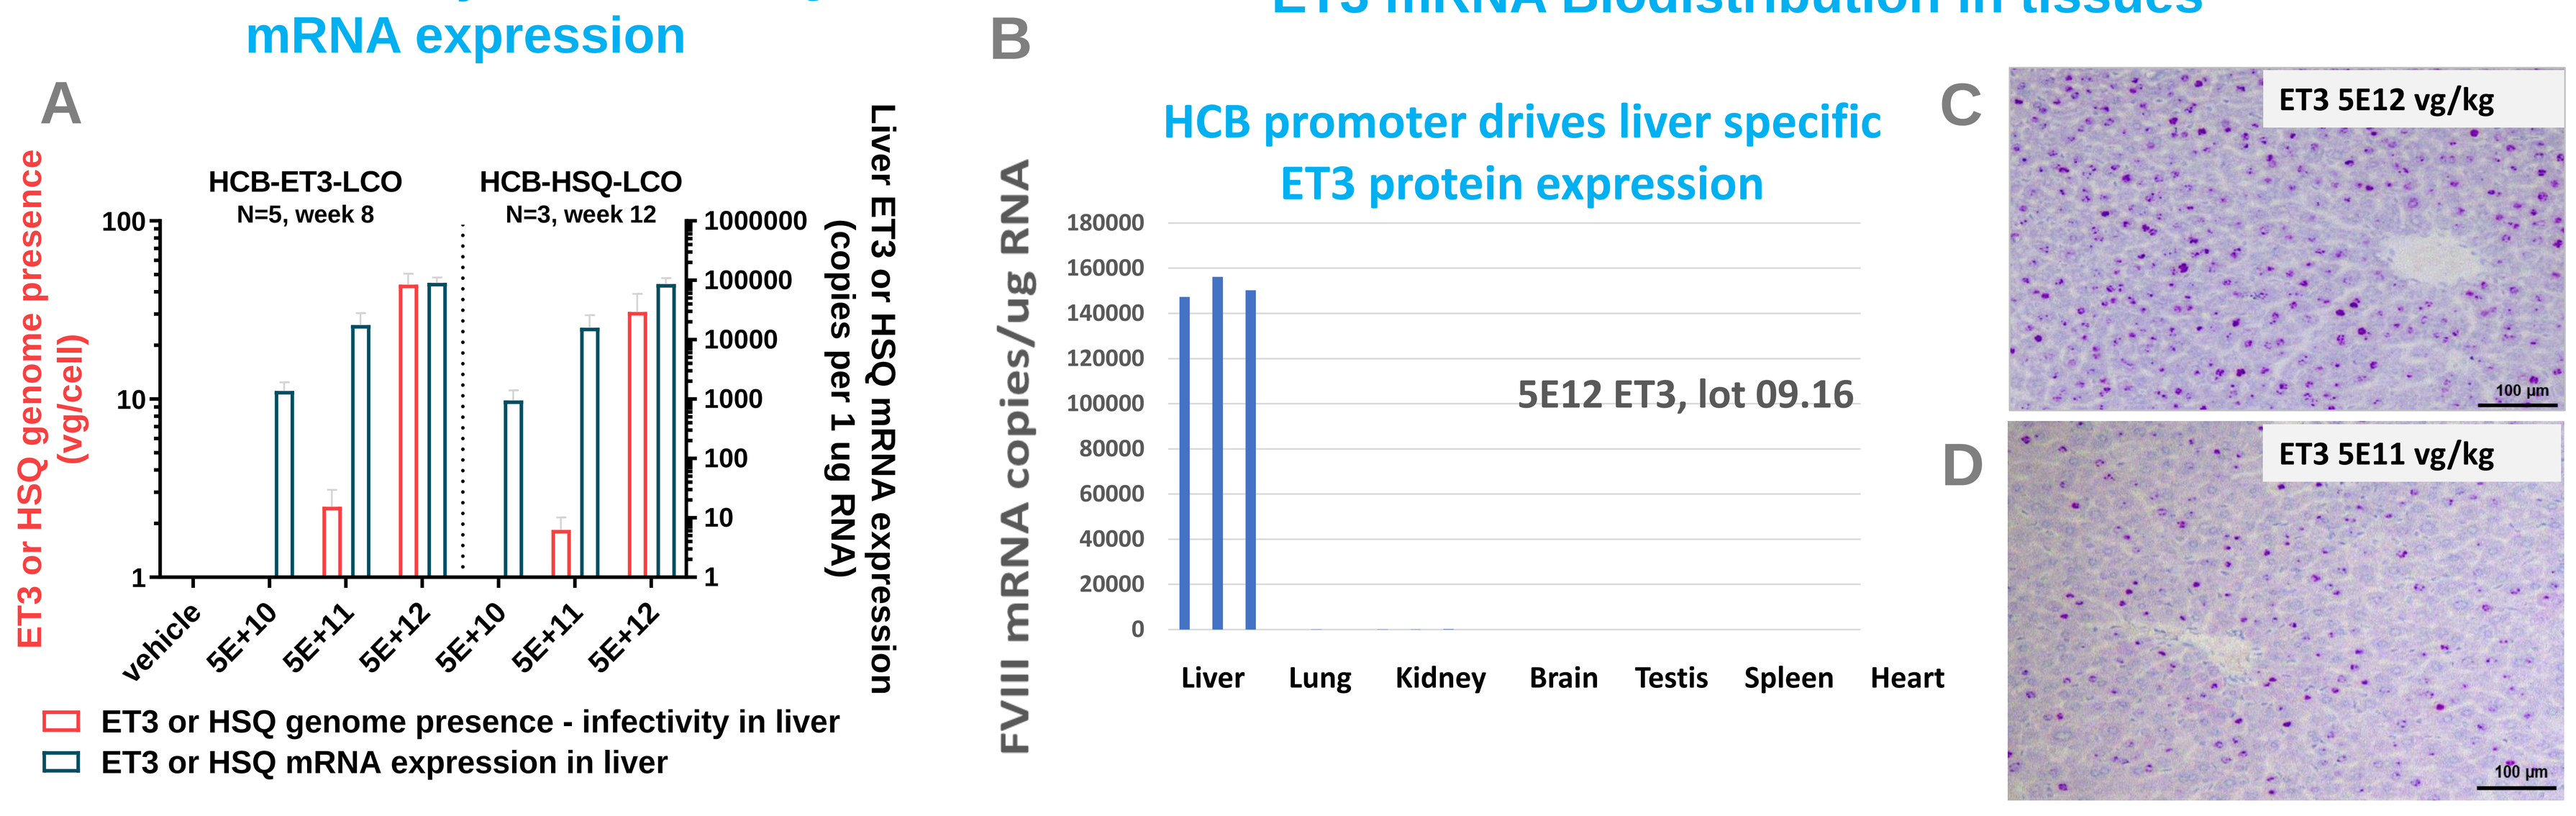

A
HCB-ET3-LCO
N=5, week 8
HCB-HSQ-LCO
N=3, week 12
ET3 or HSQ genome presence (vg/cell)
Liver ET3 or HSQ mRNA expression (copies per 1 ug RNA)
vehicle
5E+10
5E+11
5E+12
5E+10
5E+11
5E+12
ET3 or HSQ genome presence - infectivity in liver
ET3 or HSQ mRNA expression in liver
B
HCB promoter drives liver specific ET3 protein expression
FVIII mRNA copies/ug RNA
5E12 ET3, lot 09.16
Liver
Lung
Kidney
Brain
Testis
Spleen
Heart
C
ET3 5E12 vg/kg
D
ET3 5E11 vg/kg

Fig 6. A. In vivo Infectivity - ET3 or HSQ genome presence in liver tissue (ddPCR, vg/cell – 4 lobe mean, SD), liver ET3 or HSQ mRNA expression (ddPCR, copy number/µg RNA – 4 lobe mean, SD); **B.** ET3 mRNA tissue biodistribution – 5E12 vg/kg, data from 3 mice.
C, D. ET3 fVIII mRNA tissue distribution by liver RNAscope (red – ET3 mRNA); **C** - ET3 5E12 vg/kg dose; **D** - ET3 5E11 vg/kg dose.
Summary: ET3/HSQ genome liver presence and mRNA expression levels were comparable between constructs, while circulating protein levels showed clear ET3 advantage in this murine model. ET3 mRNA was specifically expressed in C57Bl/6 liver, indicating a high liver specificity of HCB promoter.

ASC-618 Efficacy In Vivo – NHP (Cynomolgus Macaque) Model

Correlation between ET3/HSQ plasma protein levels and hFVIII Inhibitors by Bethesda

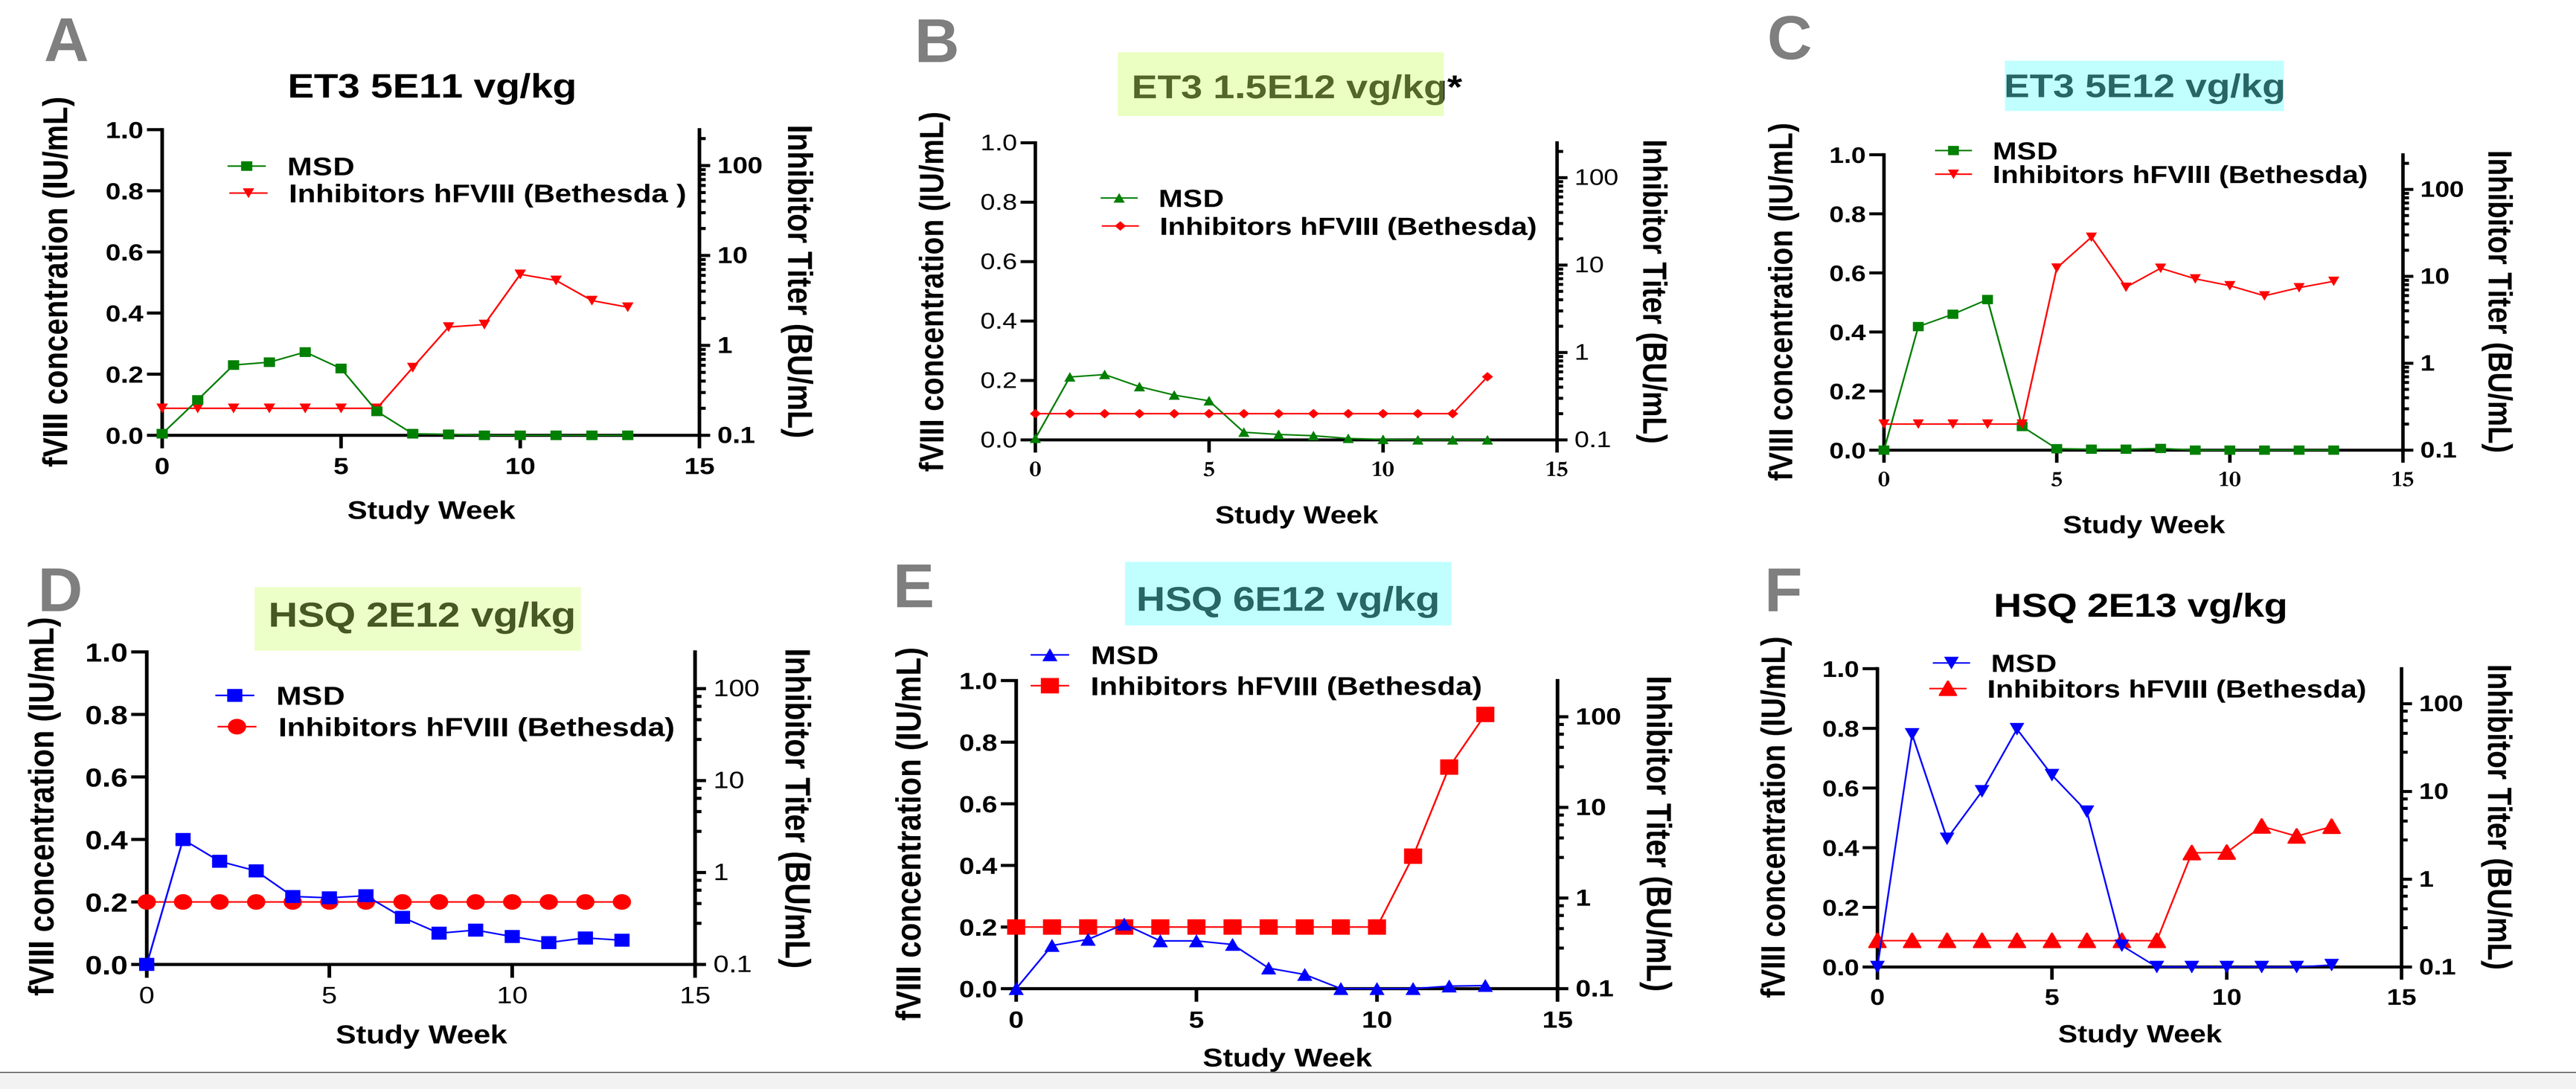

A
ET3 5E11 vg/kg
B
ET3 1.5E12 vg/kg*
C
ET3 5E12 vg/kg
D
HSQ 2E12 vg/kg
E
HSQ 6E12 vg/kg
F
HSQ 2E13 vg/kg
MSD
Inhibitors hFVIII (Bethesda)
fVIII concentration (IU/mL)
Inhibitor Titer (BU/mL)
Study Week

Fig 7. A-F. Human fVIII circulating levels in NHP plasma by MSD (green line – HCB-ET3-LCO; blue line – HCB-HSQ-LCO; IU/mL), and human fVIII inhibitors by Bethesda (red line, BU/mL) shown (n=1). Only anti-AAV8 antibody negative animals were enrolled in the study. Colored boxes allow for comparison between similar ET3 and HSQ dose levels. * Lower than expected infectivity (Fig 8.A) was observed in the ET3 1.5E12 vg/kg animal.
Summary: ET3 demonstrated efficient protein expression at the lowest 5E11 vg/kg dose. Anti-hFVIII inhibitor levels were protein expression dependent, indicating successful ET3/HSQ protein production following treatment.

Liver infectivity and ET3/HSQ mRNA expression

mRNA Biodistribution in tissues

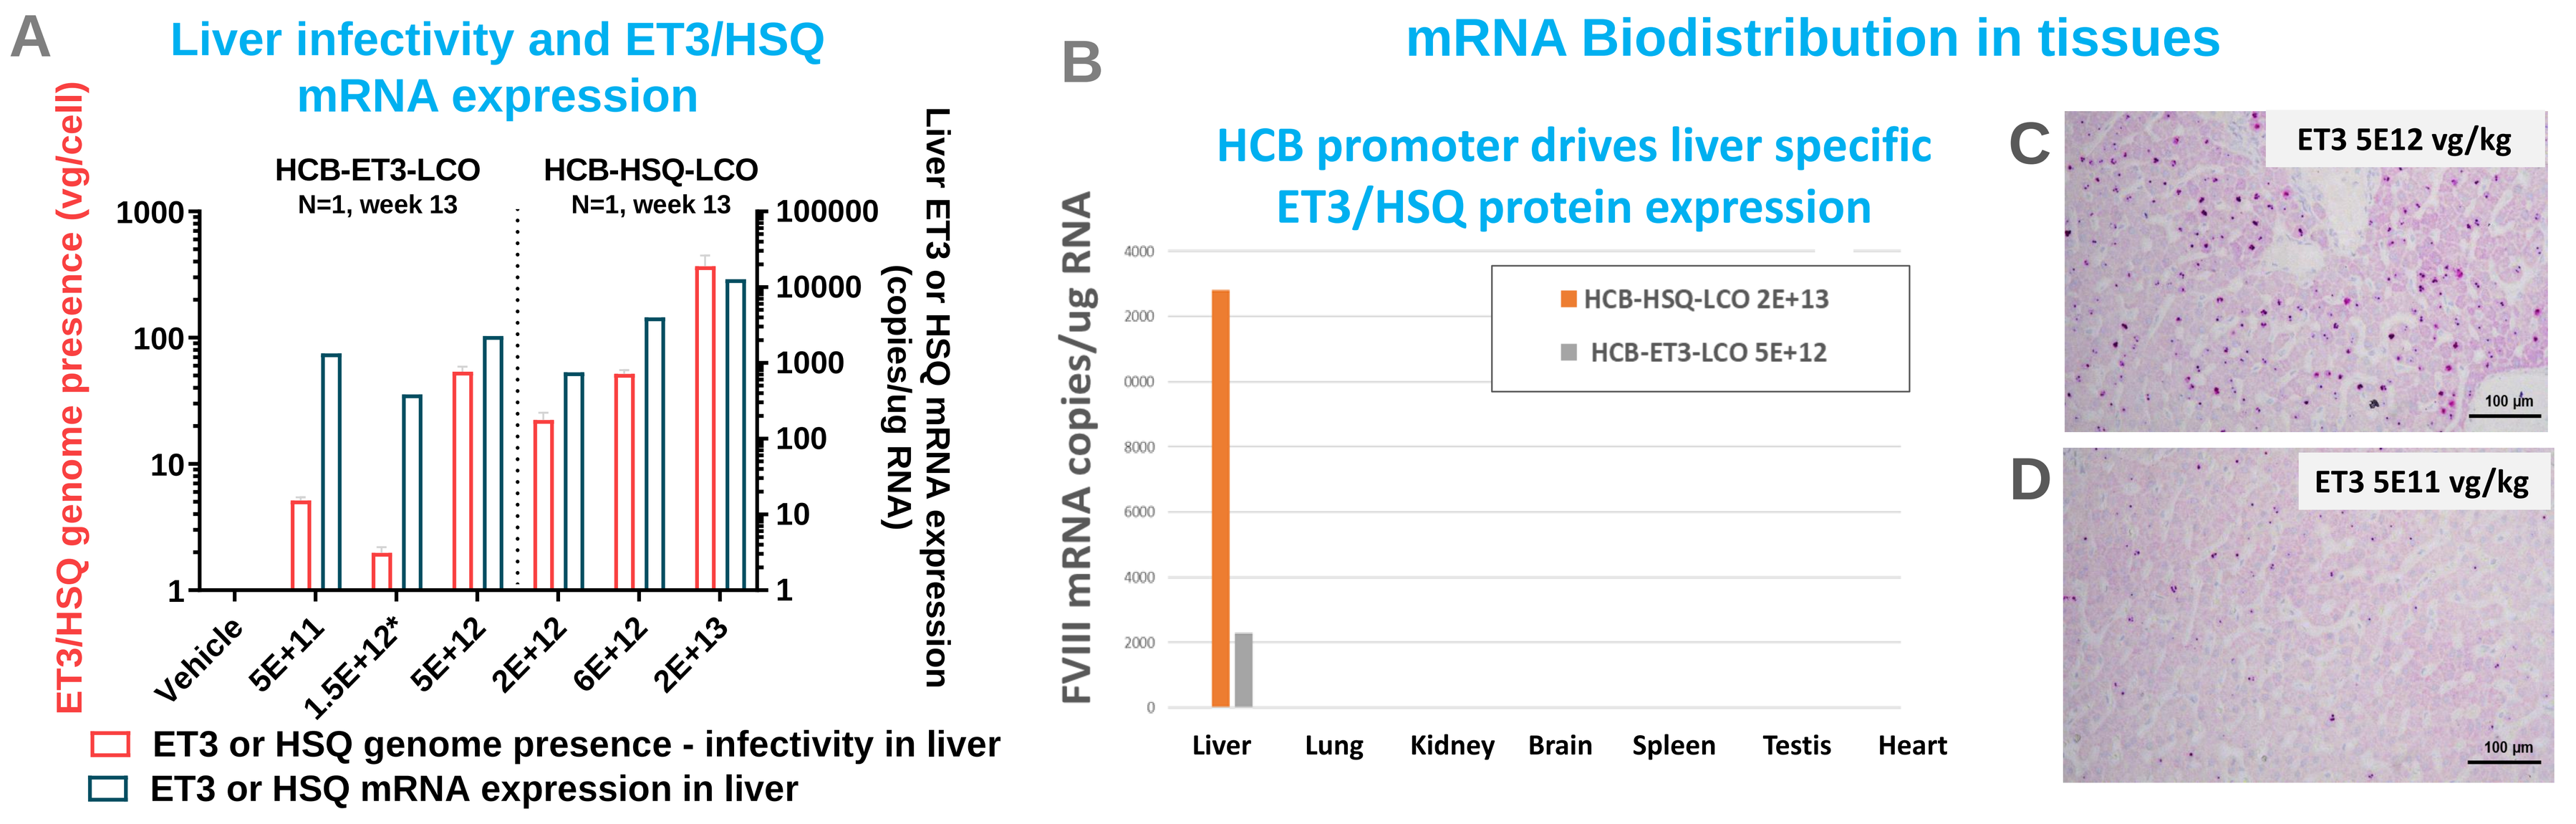

A
HCB-ET3-LCO
N=1, week 13
HCB-HSQ-LCO
N=1, week 13
ET3/HSQ genome presence (vg/cell)
Liver ET3 or HSQ mRNA expression (copies/ug RNA)
Vehicle
5E+11
1.5E+12*
5E+12
2E+12
6E+12
2E+13
ET3 or HSQ genome presence - infectivity in liver
ET3 or HSQ mRNA expression in liver
B
HCB promoter drives liver specific ET3/HSQ protein expression
FVIII mRNA copies/ug RNA
HCB-HSQ-LCO 2E+13
HCB-ET3-LCO 5E+12
Liver
Lung
Kidney
Brain
Spleen
Testis
Heart
C
ET3 5E12 vg/kg
D
ET3 5E11 vg/kg

Fig 8. A. In vivo Infectivity - ET3 or HSQ genome presence in liver tissue (ddPCR, vg/cell – 4 lobe mean, SD), ET3 mRNA expression in liver tissue (ddPCR, copy number/µg RNA, left lobe); **B**. ET3/HSQ mRNA Biodistribution in tissues (left liver lobe). **C,D**. ET3 fVIII mRNA tissue distribution by liver RNAscope (red – ET3 mRNA); **C.** 5E12 vg/kg dose (left liver lobe); **D.** 5E11 vg/kg dose (left liver lobe).
Summary: ET3 and HSQ liver infectivity and mRNA expression were generally comparable and dose dependent. mRNA expression was detected only in liver tissue, indicating HCB promoter specificity. Vector DNA biodistribution was analyzed and showed similar pattern for both constructs (data not shown).

Conclusions

- In the presented preclinical in vivo studies gene therapy vector ASC-618 (AAV8-HCB-ET3-LCO) was tested within the overall dose range spanning between 5E10 vg/kg and 5E12 vg/kg in 3 different animal models – C57Bl/6 mice, humanized liver FRGKO mice and NHPs. All administered doses were confirmed by subsequent ddPCR analysis of dose formulations
- Efficient, dose-dependent ET3 fVIII protein expression was seen in all 3 models, even at low doses administered - 5E10 vg/kg in C57Bl/6 mice, and 5E11 vg/kg in NHPs
- Enhanced fVIII biosynthesis/secretion was confirmed for ASC-618 by higher circulating fVIII protein levels compared to HSQ, while AAV infectivity as well as fVIII mRNA expression in all 3 models were comparable for both constructs
- Inhibitors against human fVIII were developed in NHPs with ASC-618 and HSQ treatment. Inhibitor levels were protein expression dependent, indicating successful fVIII protein production following ASC-618 treatment
- ASC-618 and AAV8-HCB-HSQ-LCO showed similar viral DNA biodistribution across multiple organs in NHPs
- FVIII mRNA was specifically expressed in mouse and NHP liver, indicating a high liver specificity of HCB promoter
- No safety concerns or differences compared to vehicle controls were observed in ASC-618 treated mice and NHPs, including body weight, body temperature, clinical signs, liver enzymes, hematological parameters and histopathology
- These preclinical studies will inform dose selection for first in human ASC-618 gene therapy for hemophilia A

Acknowledgements

We would like to thank Alex Lyubimov, Kasim Kabirov and Matt Lindeblad from University of Illinois in Chicago, Todd Haryu from PreLabs, and Lander Foquet from Yecuris for their support with animal study design and implementation. We also thank Yongmei Luo Chen from Center for Biomedical Testing, Inc. for her support in assay development and study sample testing.